Alpha 1 Anti-Trypsin (AAT) offers potent therapy for Steroid Resistant Gut GVHD: Interim Results of a Phase I/II Clinical Study

A.Mario Marcondes[1,2] Terry Fulong[1], Mohamed L. Sorror[1,2], Ekapun Karoopongse[1], Marina Lesnikova[1], Sabina Janciauskiene[3], Charles A Dinarello[4] , David Hockenbery[1,2] and H. Joachim Deeg[1,2]

1) Clinical Research Division, Fred Hutchinson Cancer Research Center, 2) Division of Oncology, University of Washington, Seattle, Washington 3) Respiratory Medicine, Hannover Medical School, Hannover, Germany 4) Department of Medicine, University of Colorado Denver, Aurora, Colorado.

Abstract:

Background and Hypothesis: Graft-versus-host disease (GVHD) is a major complication of allogeneic hematopoietic cell transplantation (HCT). Severe acute GVHD of the gastrointestinal tract can be life threatening and is associated with a loss of acute phase proteins including alpha1-antitrypsin (AAT). AAT is a 52-kDa serum serine-protease inhibitor that is normally produced by hepatocytes and mononuclear phagocytes. AAT has anti-inflammatory properties. For example, AAT inhibits the lipopolysaccharide (LPS)-induced production and release of both TNF-α and IL-1β and enhance IL-10 production in human monocytes. In whole blood cultures, AAT inhibits the expression of IL-8, IL-6, TNF-α, and IL-1β. Moreover, AAT suppresses the activation of nuclear factor (NF)-κB, a transcription factor associated with the expression of pro-inflammatory cytokines. We and others have shown in allogeneic murine models that administration of AAT during HCT suppresses serum levels of pro-inflammatory cytokines, interferes with the development of GVHD and reduces transplant-related mortality. We embarked upon a phase I/II clinical study to evaluate the feasibility and safety of AAT given to HCT recipients with steroid non-responsive acute GVHD. We aimed at characterizing pharmacodynamic effects of AAT and assess clinical responses as determined by changes in stool volume and overall clinical improvement.

Materials And Methods:

Methods: Seven patients (3 female, 4 male) with hematologic malignancies have so far been enrolled in the first and second cohort. Patients were 35-59 (median 50.4) years old and were given transplants from HLA-matched siblings (n=5) or received cord blood (n=2) following high intensity (myeloablative) or reduced intensity conditioning with cyclophosphamide +TBI (n=2), Fludarabine+TBI (n=4), or busulfan +cyclophosphamide (n=1). All patients received cyclosporine and MMF for GVHD prophylaxis. Acute GVHD of grades III-IV developed at 49 to 71 (median of 48) days, and treatment with systemic methylprednisolone, 2 mg/kg/day was instituted. Patients showing no clinically satisfactory responses after 5 days were given AAT (GlassiaTM) at 90 mg/kg iv on day 1, followed by 30 mg/kg (first cohort) or 60 mg/kg (second cohort) every other day for a total of 8 doses (15 days).

Table 1: Clinical Parameters in AAT-Treated Subjects

Patient ID	Diagnosis	Age	Donor Type	Conditioning Regimen	GVHD prophylaxis	GVHD onset	Follow-up (m)	GVHD (I-IV)	GVHD site	AAT Dose	Response
JJC001 (004)	CMML	35	PBSC	Cy/TBI	CSA/MMF	D+49	6	III	Liver/Gut	90-30 mg/kg	PR (deceased)
BAS002 (005)	CLL	58	PBSC	Flu/TBI	CSA	D+221	6	III	Gut	90-30 mg/kg	CR
DM003 (006)	AML	50	PBSC	Bu/Cy	CSA/MMF	D+71	1	III	Gut	90-30 mg/kg	CR
WC004 (007)	NHL	59	PBSC	Flu/TBI	CSA/MMF/Siro	D+37	1	IV	Gut	90-30 mg/kg	CR (CMV/Perf)
JW005 (008)	ALL	50	MMUCB	Flu/Cy/Thi/TBI	CSP/MMF	D+34	1	IV	Liver/Gut	90-30 mg/kg	PR (deceased)
ML006 (009)	AML	58	PBSC	Clo/TBI	CSA/MMF	D+93	2	III	Gut	90-30 mg/kg	CR (ARDS/CMV/Perf)
XX007 (010)	T-ALL	35	MMUCB	Flu/Cy/TBI	CSA/MMF	D+70	1	III	Liver/Gut	90-60 mg/kg	PR

Results:

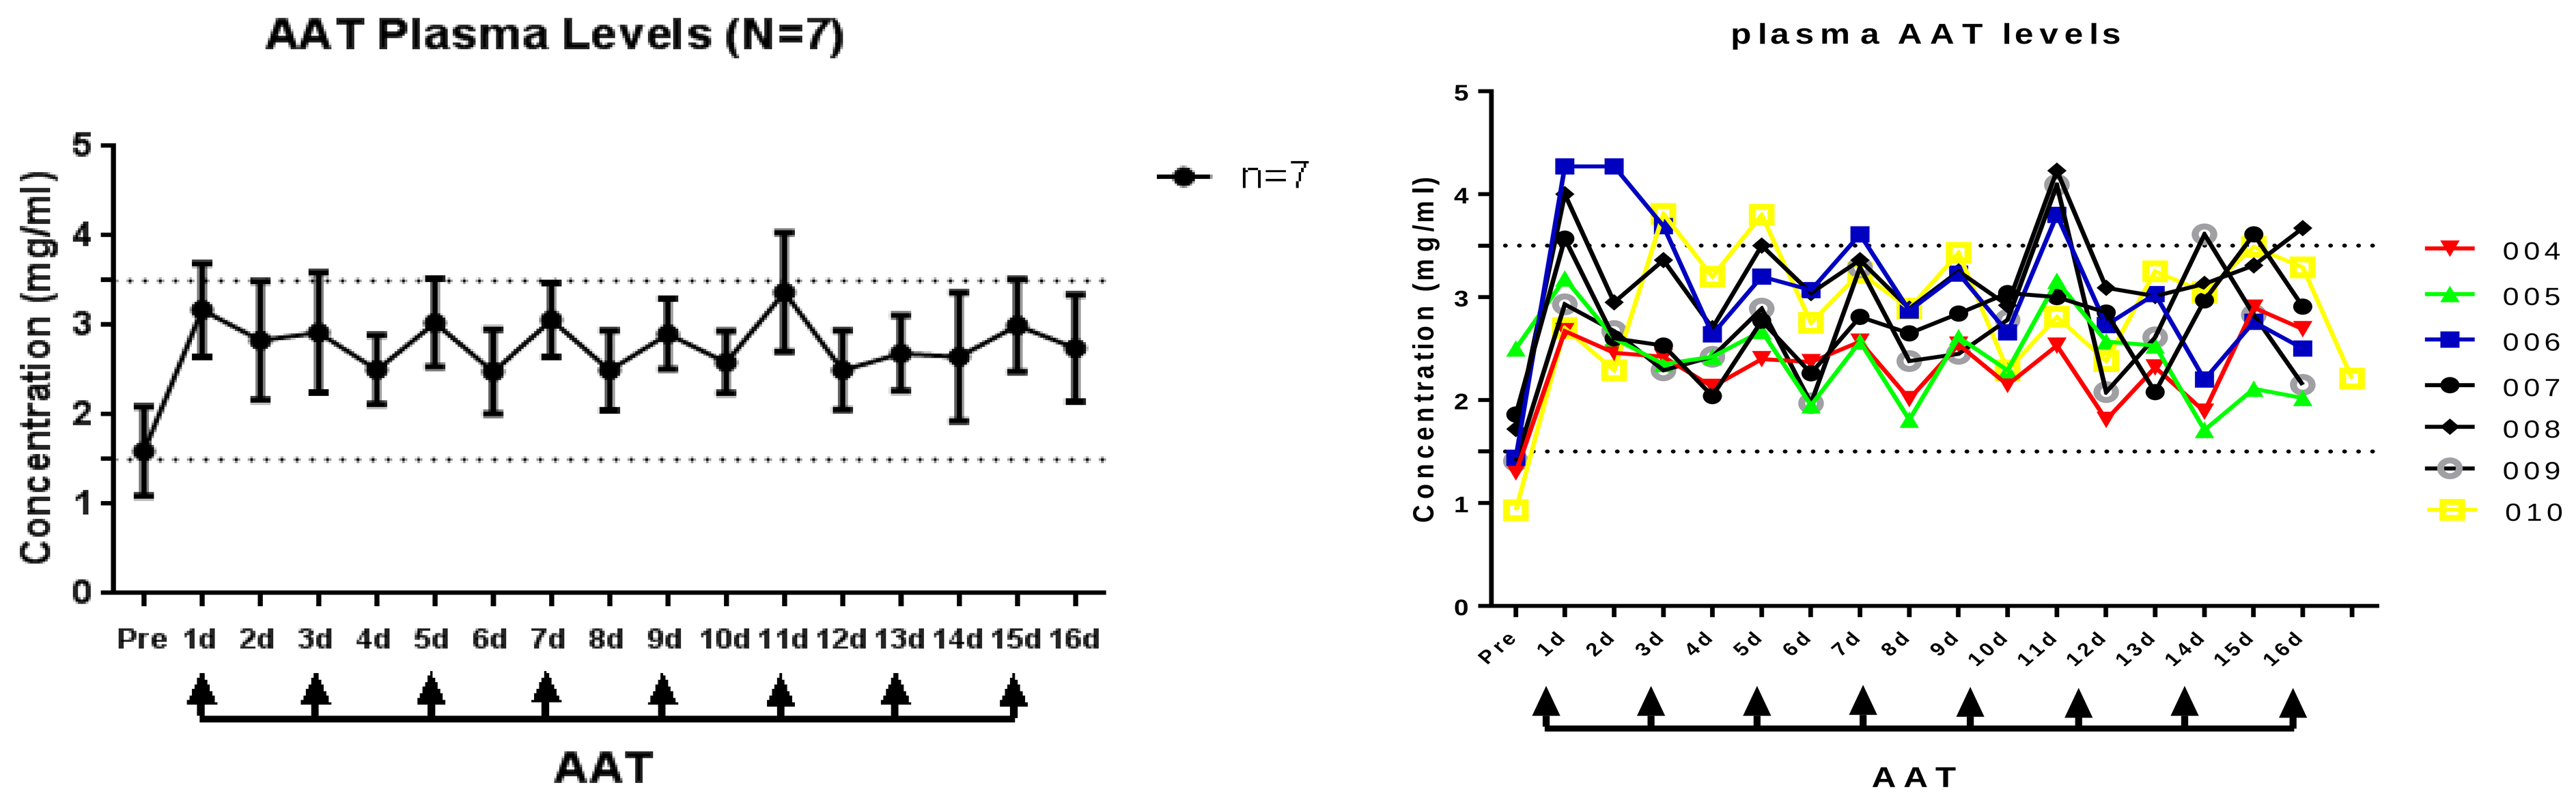

AAT Plasma Levels (N=7)
n=7
Concentration (mg/ml)
AAT
plasma AAT levels
Concentration (mg/ml)
004
005
006
007
008
009
010
AAT

Figure 1- Plasma AAT levels in human allogeneic transplant recipients . AAT levels were determined by enzyme-linked immunosorbent assay (ELISA) using 96-well polystyrene plates (Costar) coated overnight at 4° C with 0.5 μg/mL mouse anti-human AAT (R&D Systems) levels pre and post AAT therapy.

Figure 2 – AAT Stool levels and Clearance of AAT during AAT Therapy

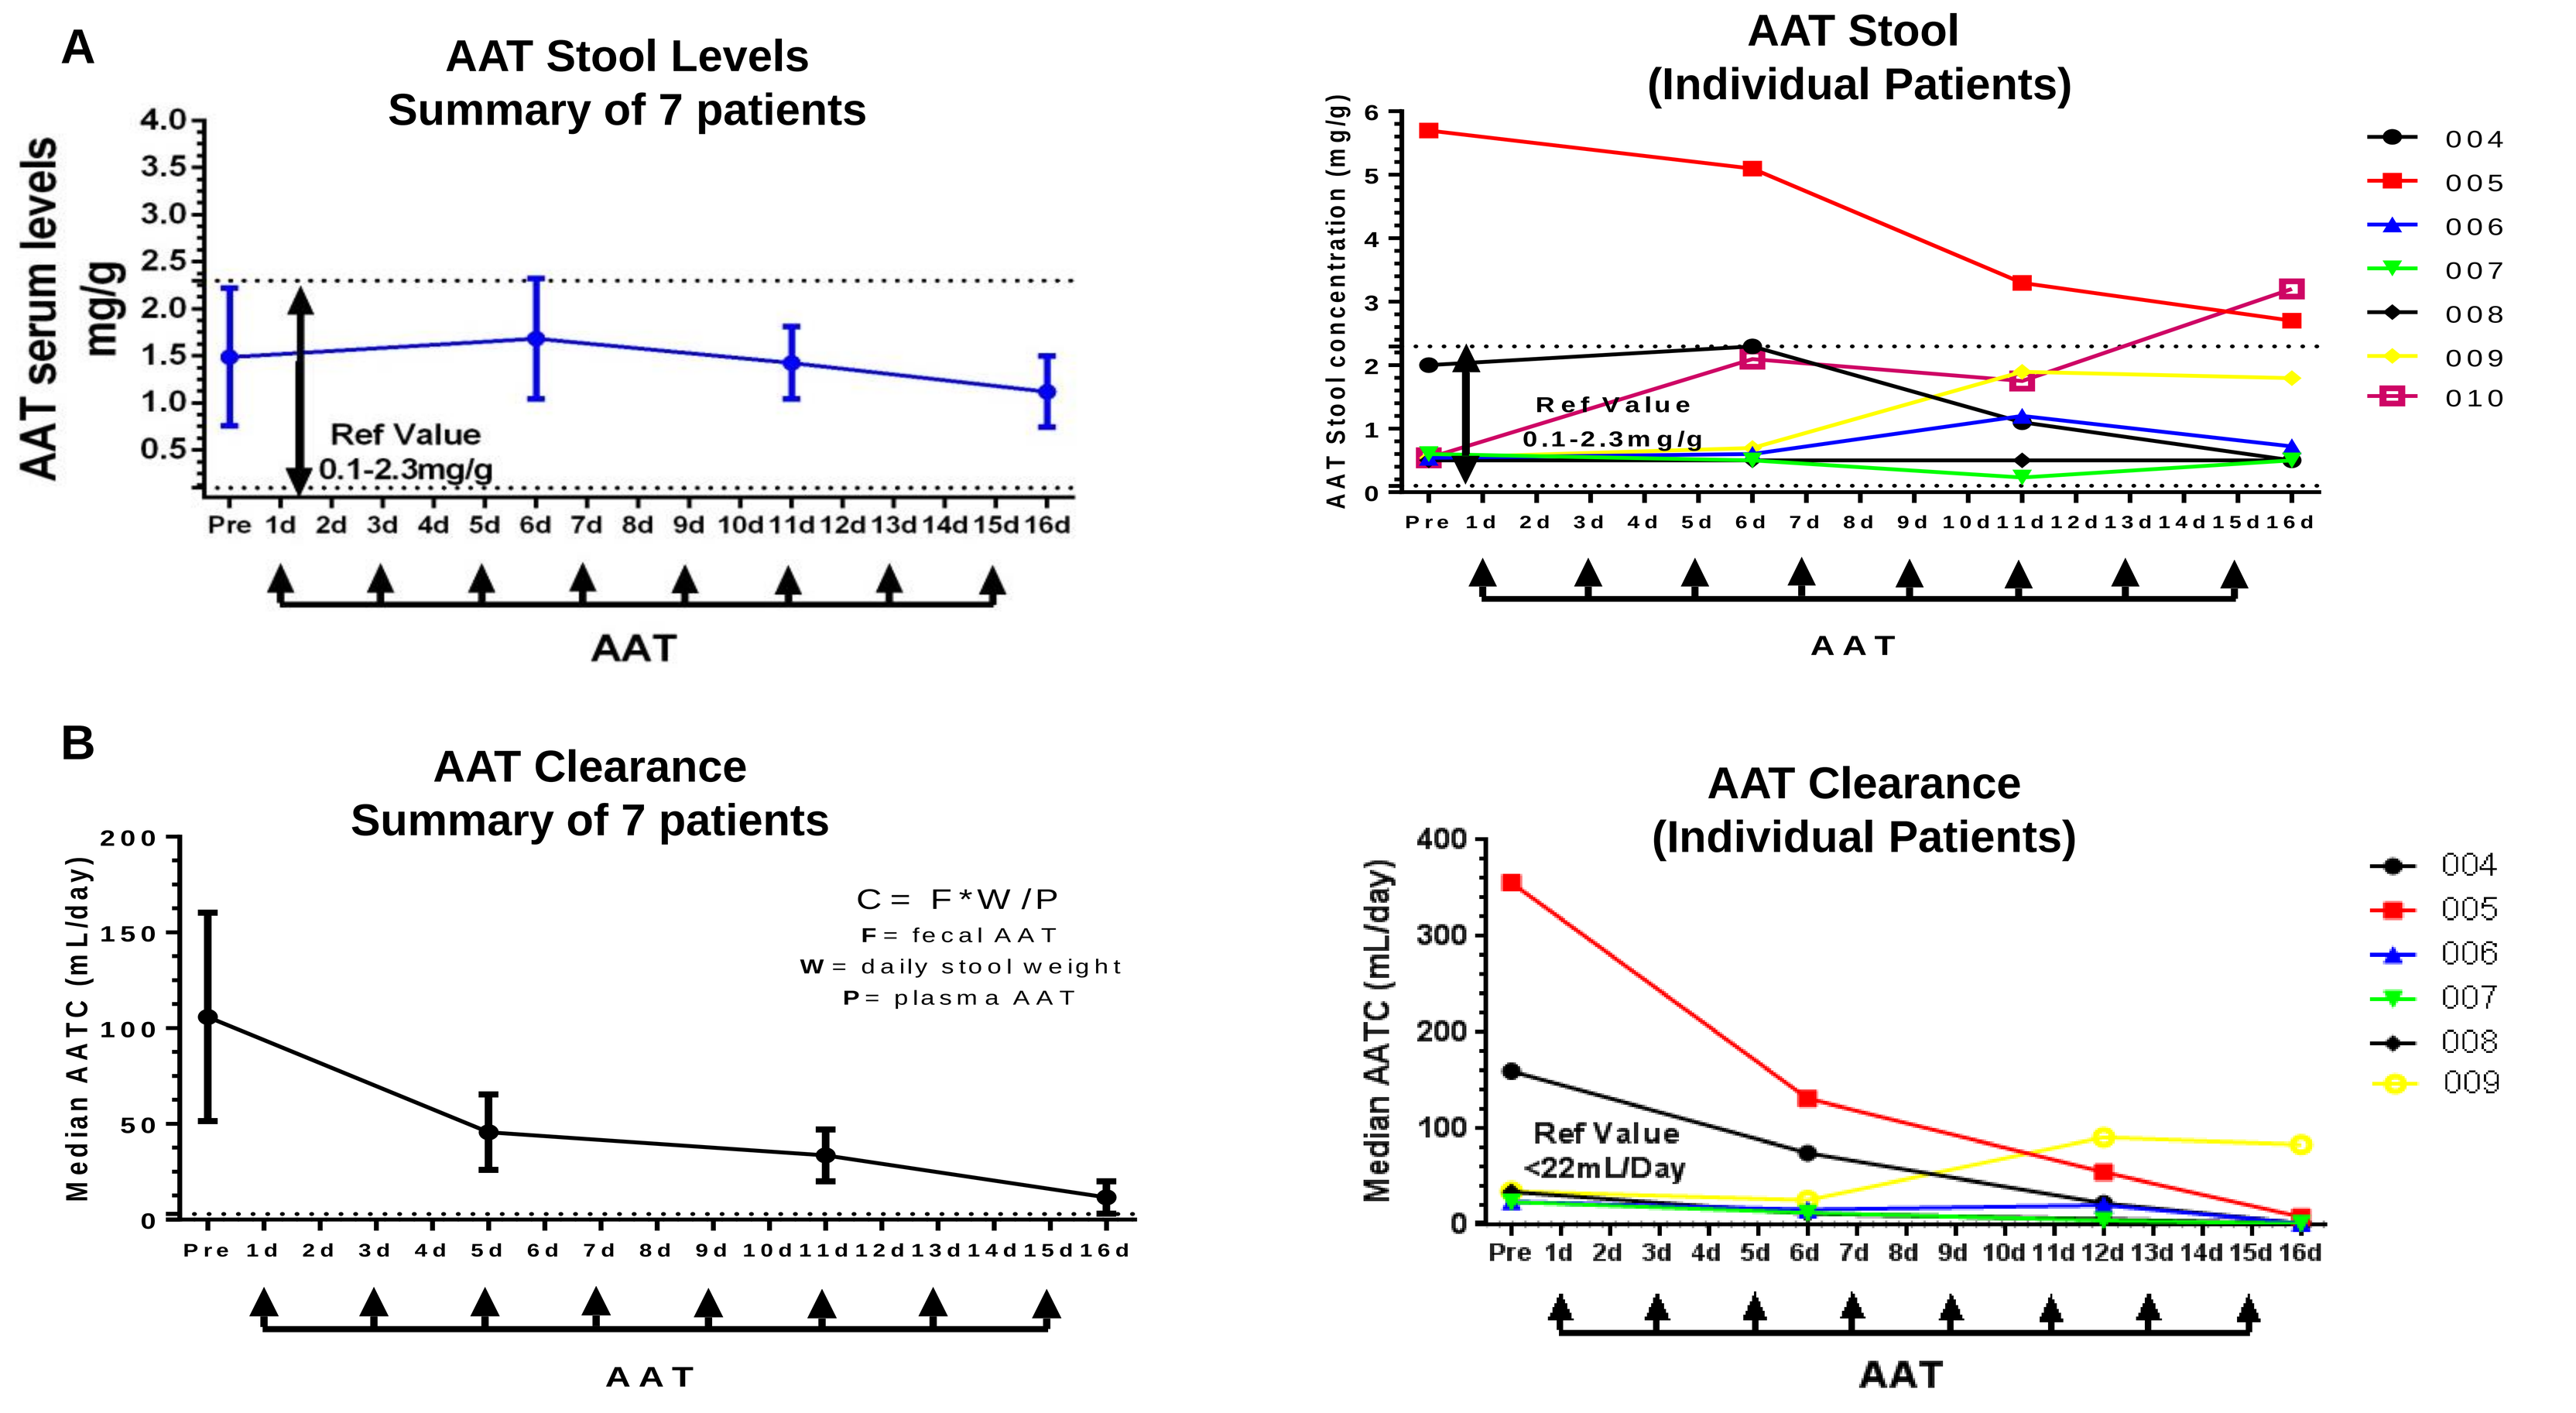

A
AAT Stool Levels
Summary of 7 patients
AAT serum levels mg/g
Ref Value
0.1-2.3mg/g
AAT
AAT Stool
(Individual Patients)
AAT Stool concentration (mg/g)
Ref Value
0.1-2.3mg/g
004
005
006
007
008
009
010
AAT
B
AAT Clearance
Summary of 7 patients
Median AATC (mL/day)
C = F*W/P
F= fecal AAT
W= daily stool weight
P= plasma AAT
AAT
AAT Clearance
(Individual Patients)
Median AATC (mL/day)
Ref Value
<22mL/Day
004
005
006
007
008
009
AAT

Figure 2 – (A) Stool AAT content per gram of stool following AAT therapy. **(B)** AAT clearance test ; AAT clearance is calculated as C = F*W/P, where C = clearance, F = fecal alpha-1-antitrypsin concentration (mg/gm), W = daily stool weight, and p = serum concentration (mg/dL). The cut-off value that the determines alpha-1-antitrypsin clearance using a nephelometric assay, and levels >22 mL/day indicate the diagnosis of Protein Loosing Enteropathy.

Figure 3 – Cytokine changes during AAT therapy

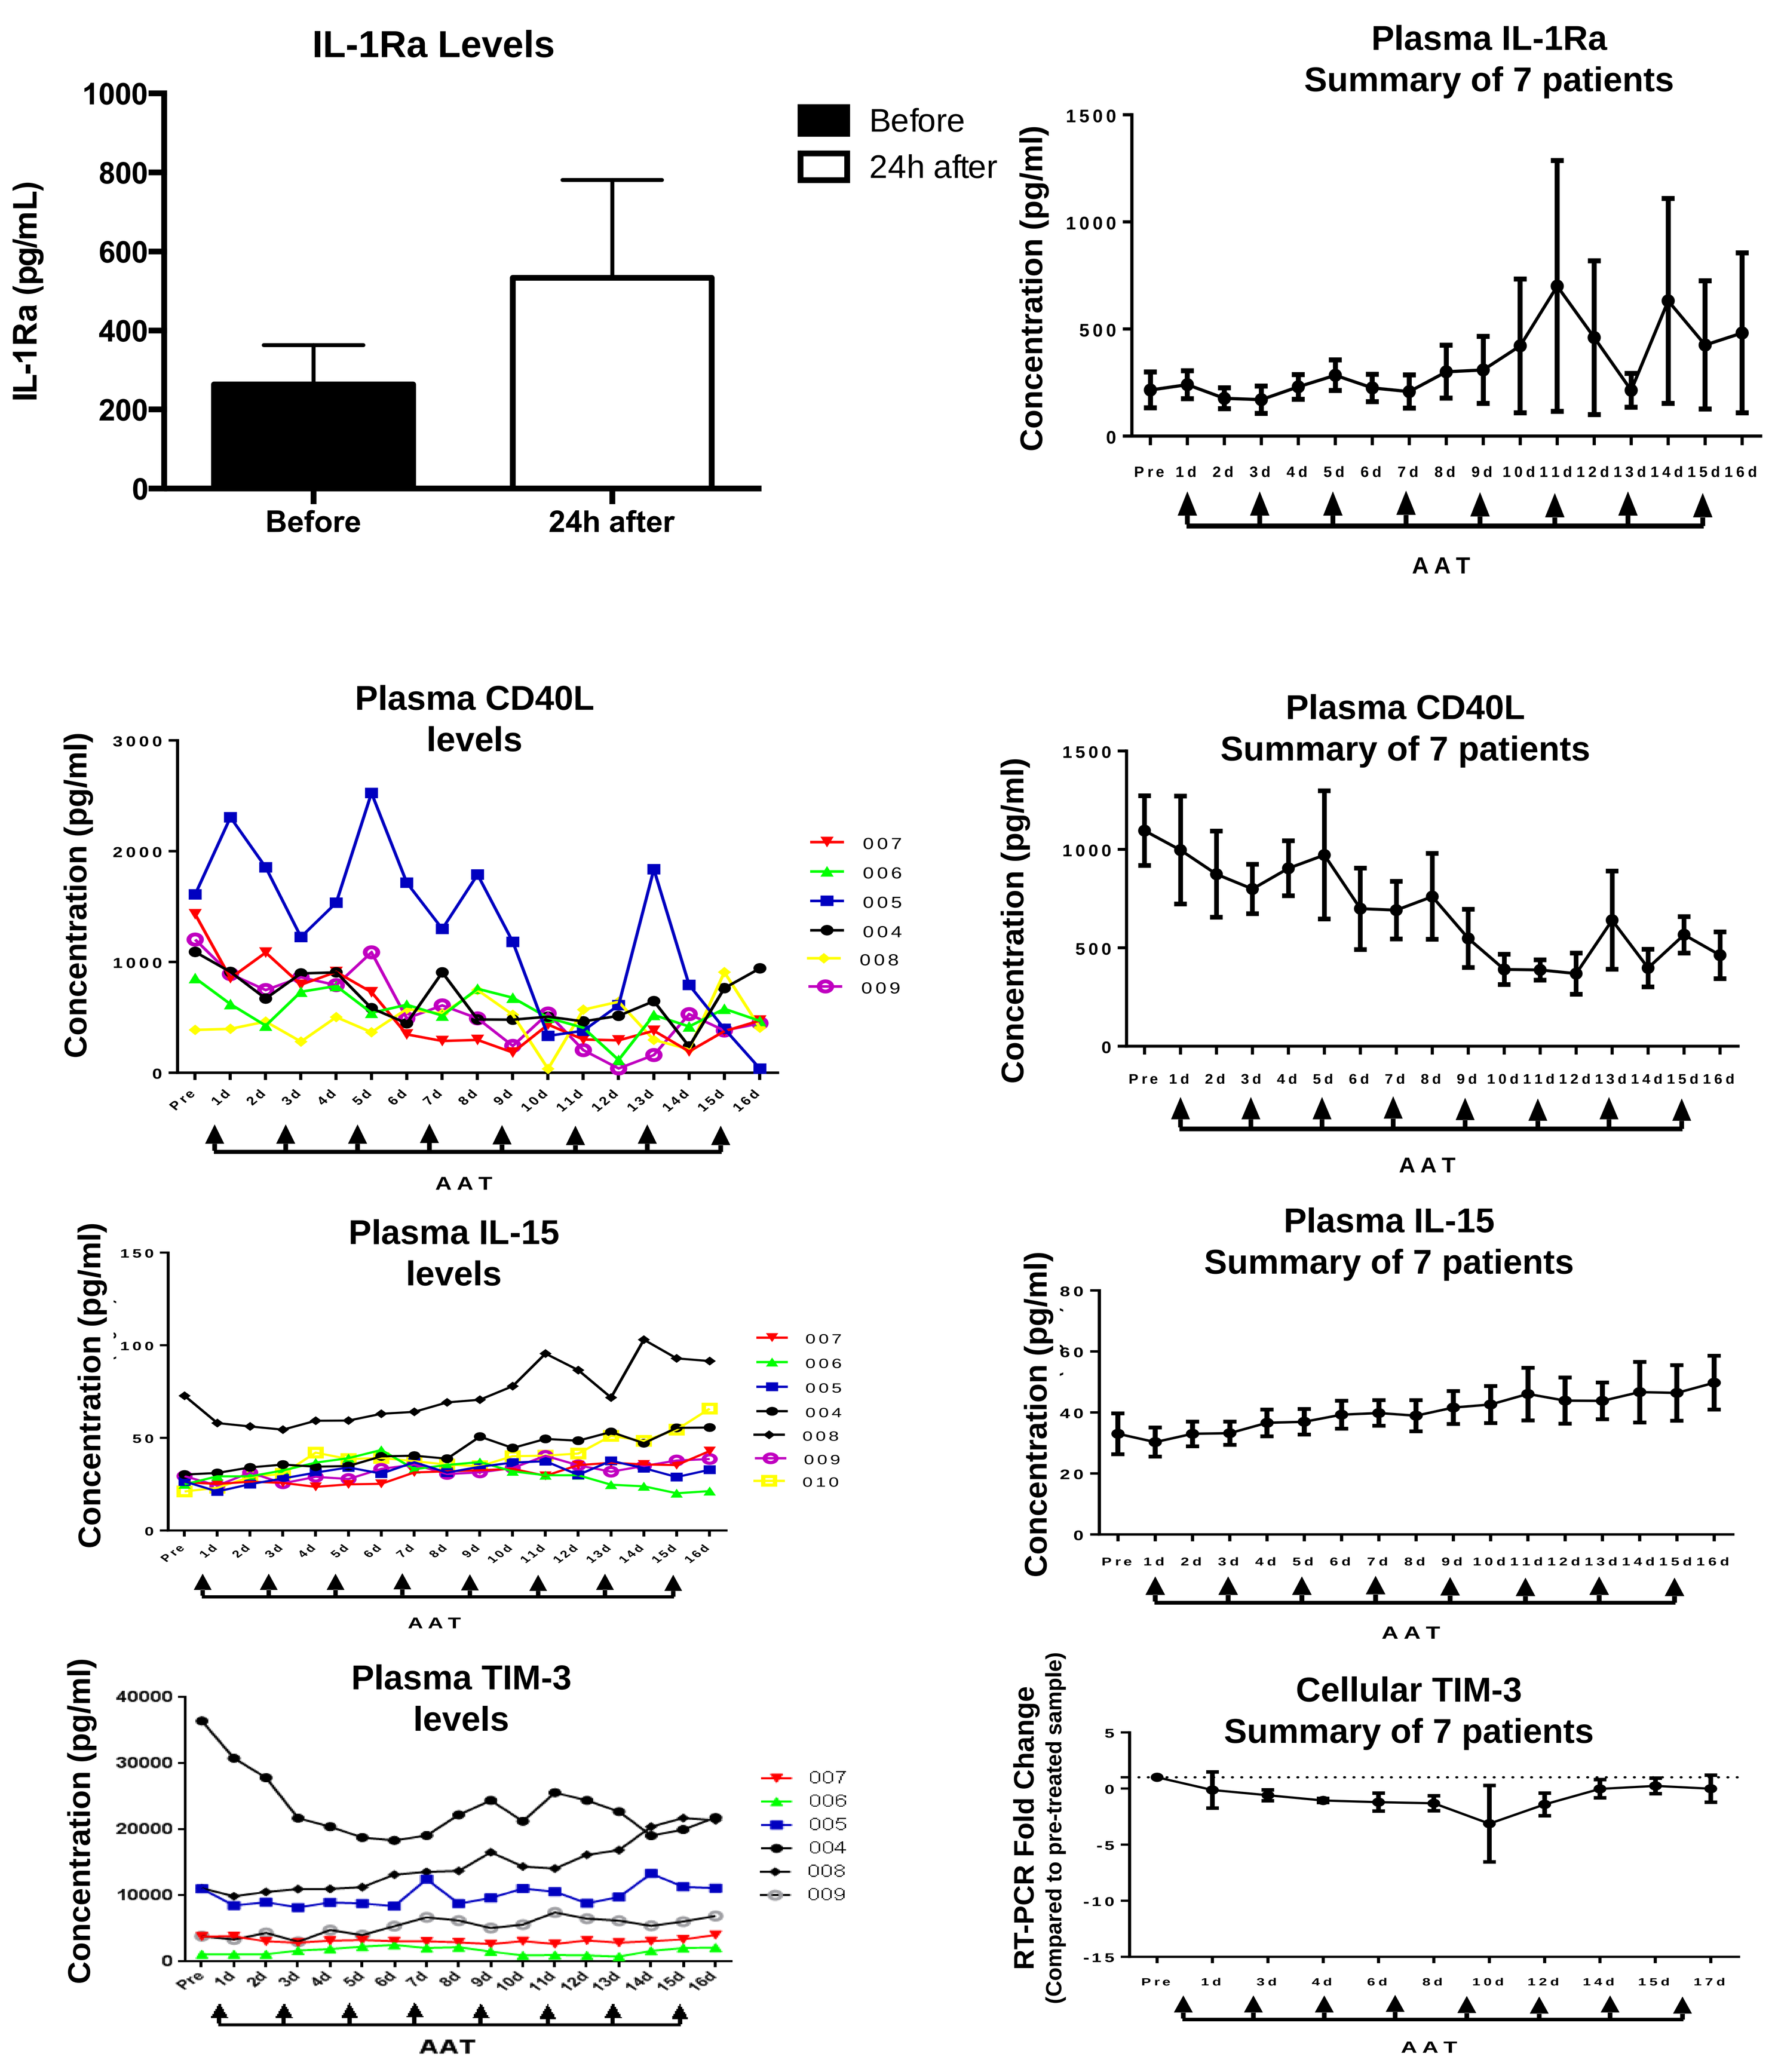

IL-1Ra Levels
IL-1Ra (pg/mL)
Before
24h after
Plasma IL-1Ra
Summary of 7 patients
Concentration (pg/ml)
AAT
Plasma CD40L
levels
Concentration (pg/ml)
007
006
005
004
008
009
AAT
Plasma CD40L
Summary of 7 patients
Concentration (pg/ml)
AAT
Plasma IL-15
levels
Concentration (pg/ml)
007
006
005
004
008
009
010
AAT
Plasma IL-15
Summary of 7 patients
Concentration (pg/ml)
AAT
Plasma TIM-3
levels
Concentration (pg/ml)
007
006
005
004
008
009
AAT
Cellular TIM-3
Summary of 7 patients
RT-PCR Fold Change
(Compared to pre-treated sample)
AAT

Figure 3 – ELISA Plasma IL-1Ra, CD40L, IL-15 and TIM-3 responses during AAT therapy . The bars indicate the mean values for the cohort of patients.

Figure 4- Endoscopic Findings (UGI)

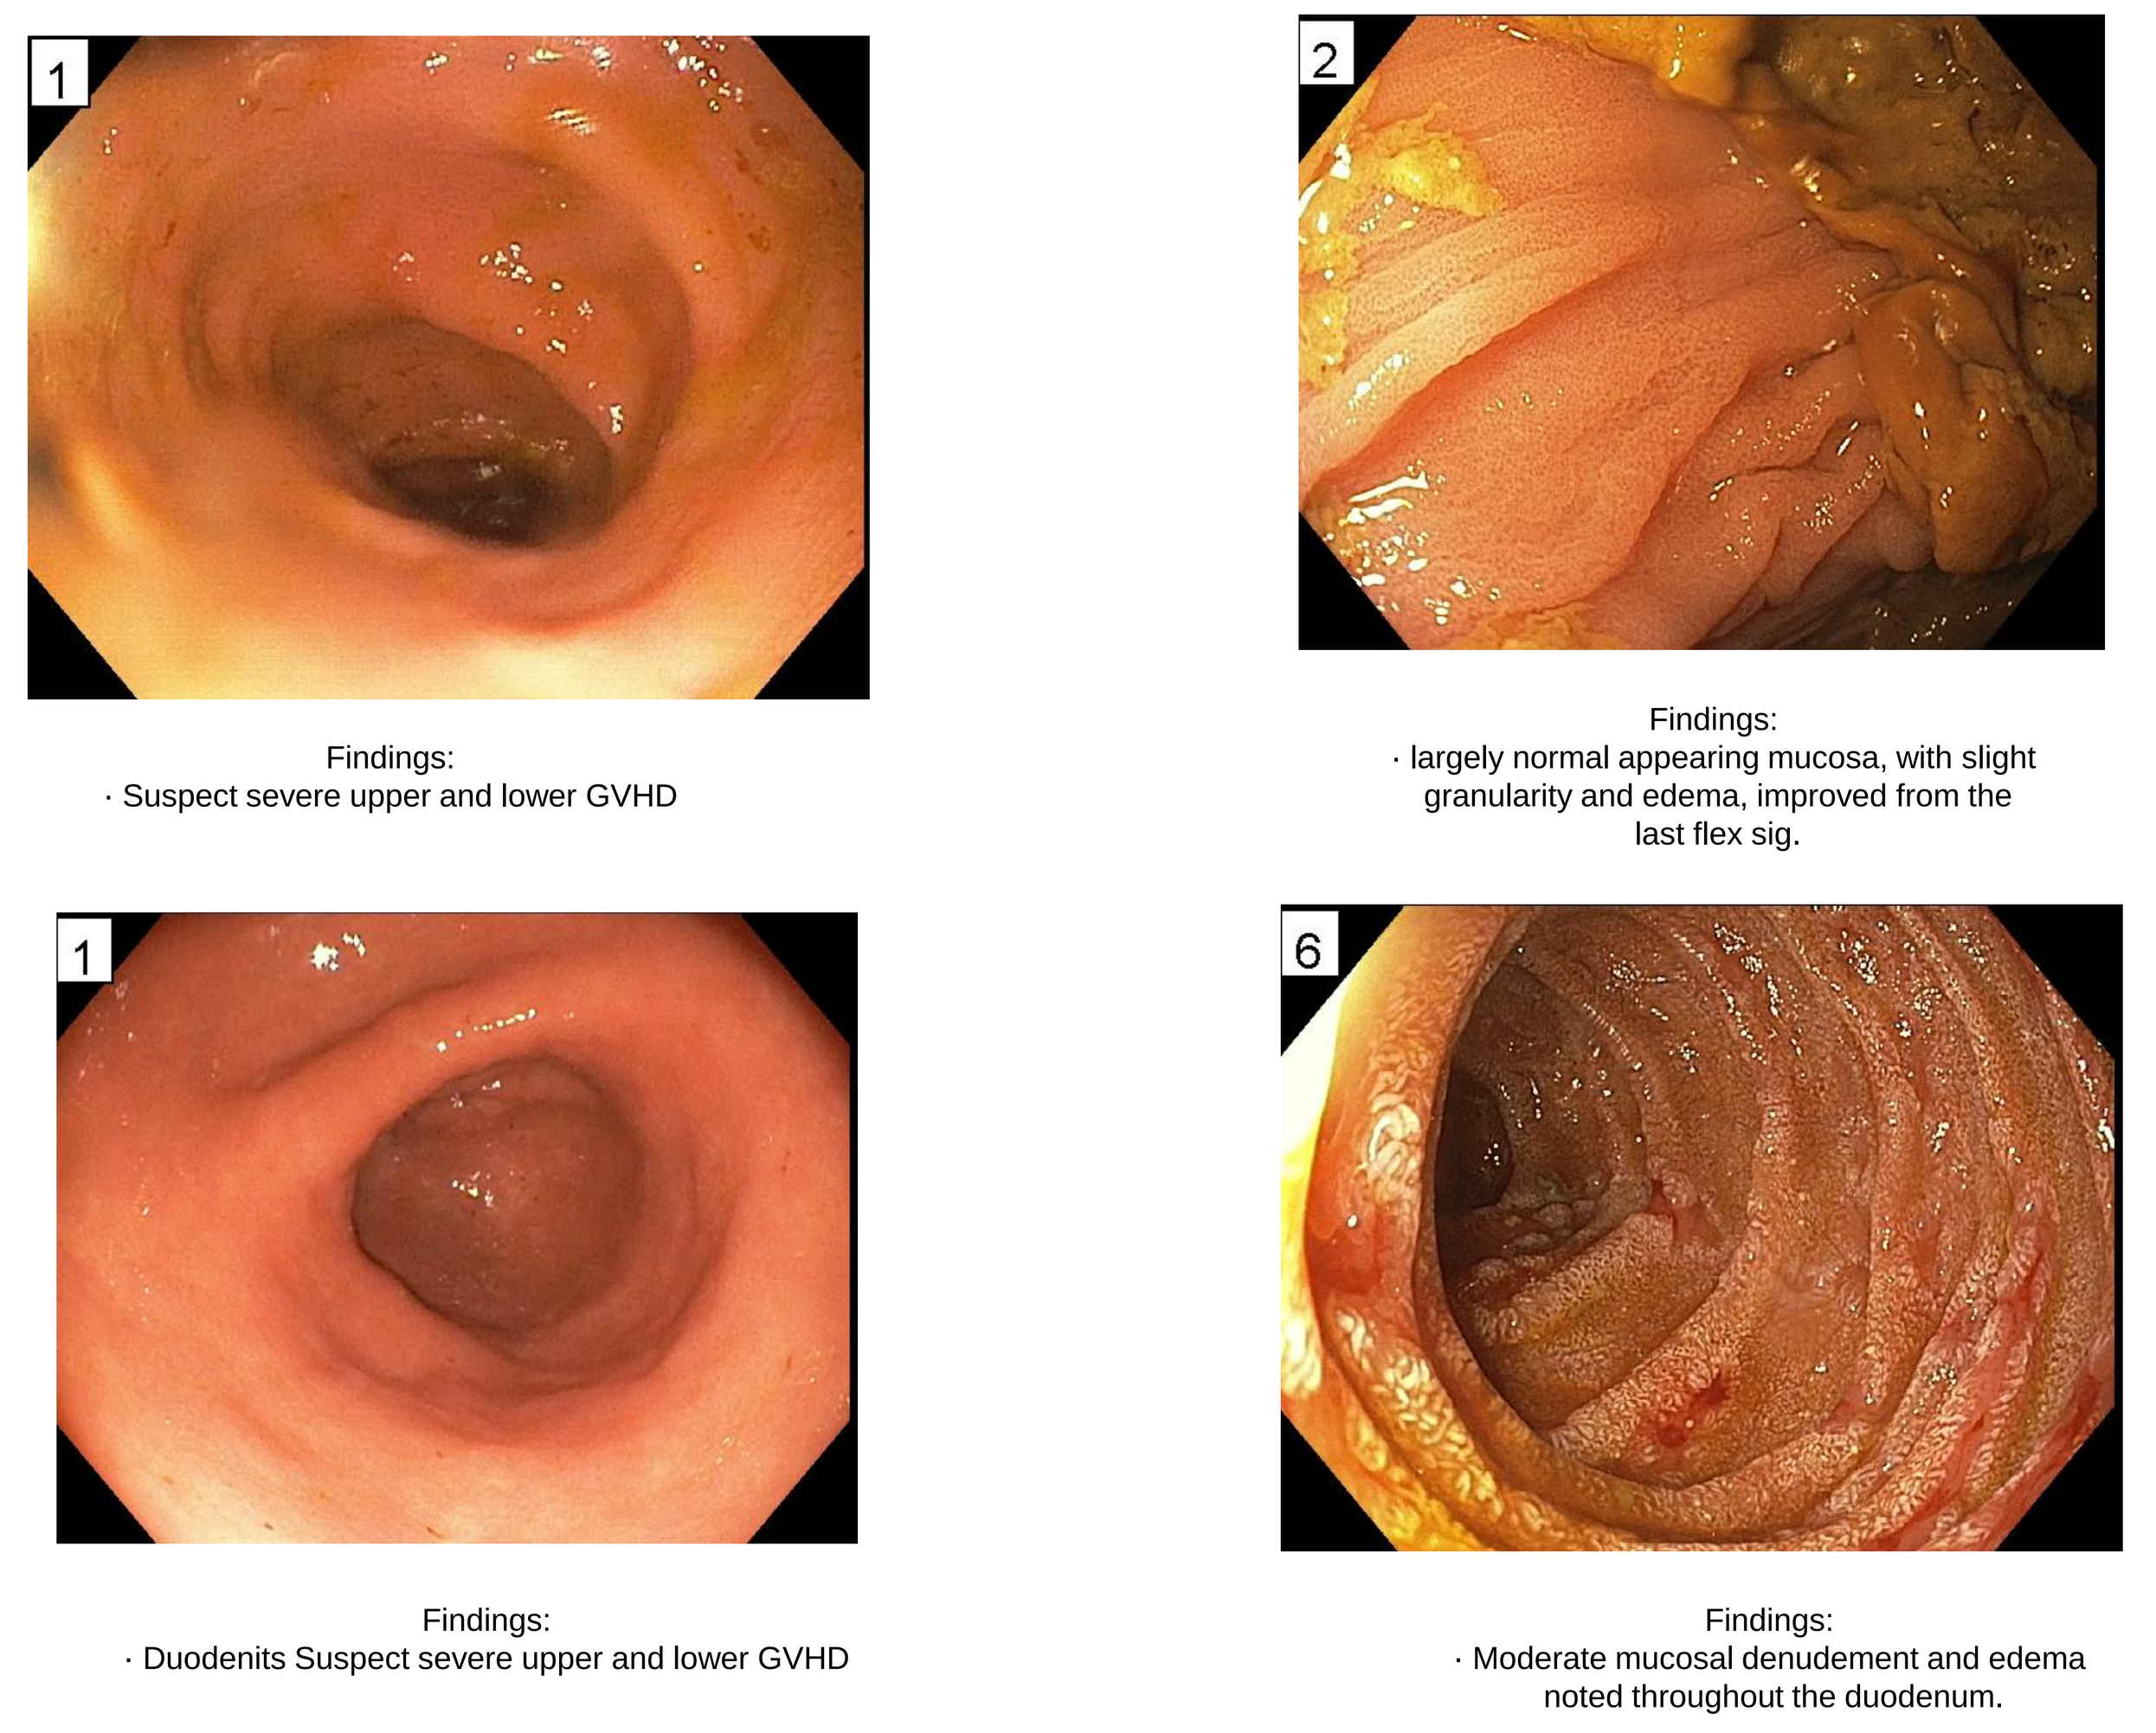

1
Findings:
· Suspect severe upper and lower GVHD
2
Findings:
· largely normal appearing mucosa, with slight granularity and edema, improved from the last flex sig.
1
Findings:
· Duodenits Suspect severe upper and lower GVHD
6
Findings:
· Moderate mucosal denudement and edema noted throughout the duodenum.

Figure 5- FACS Analysis pre and post AAT therapy

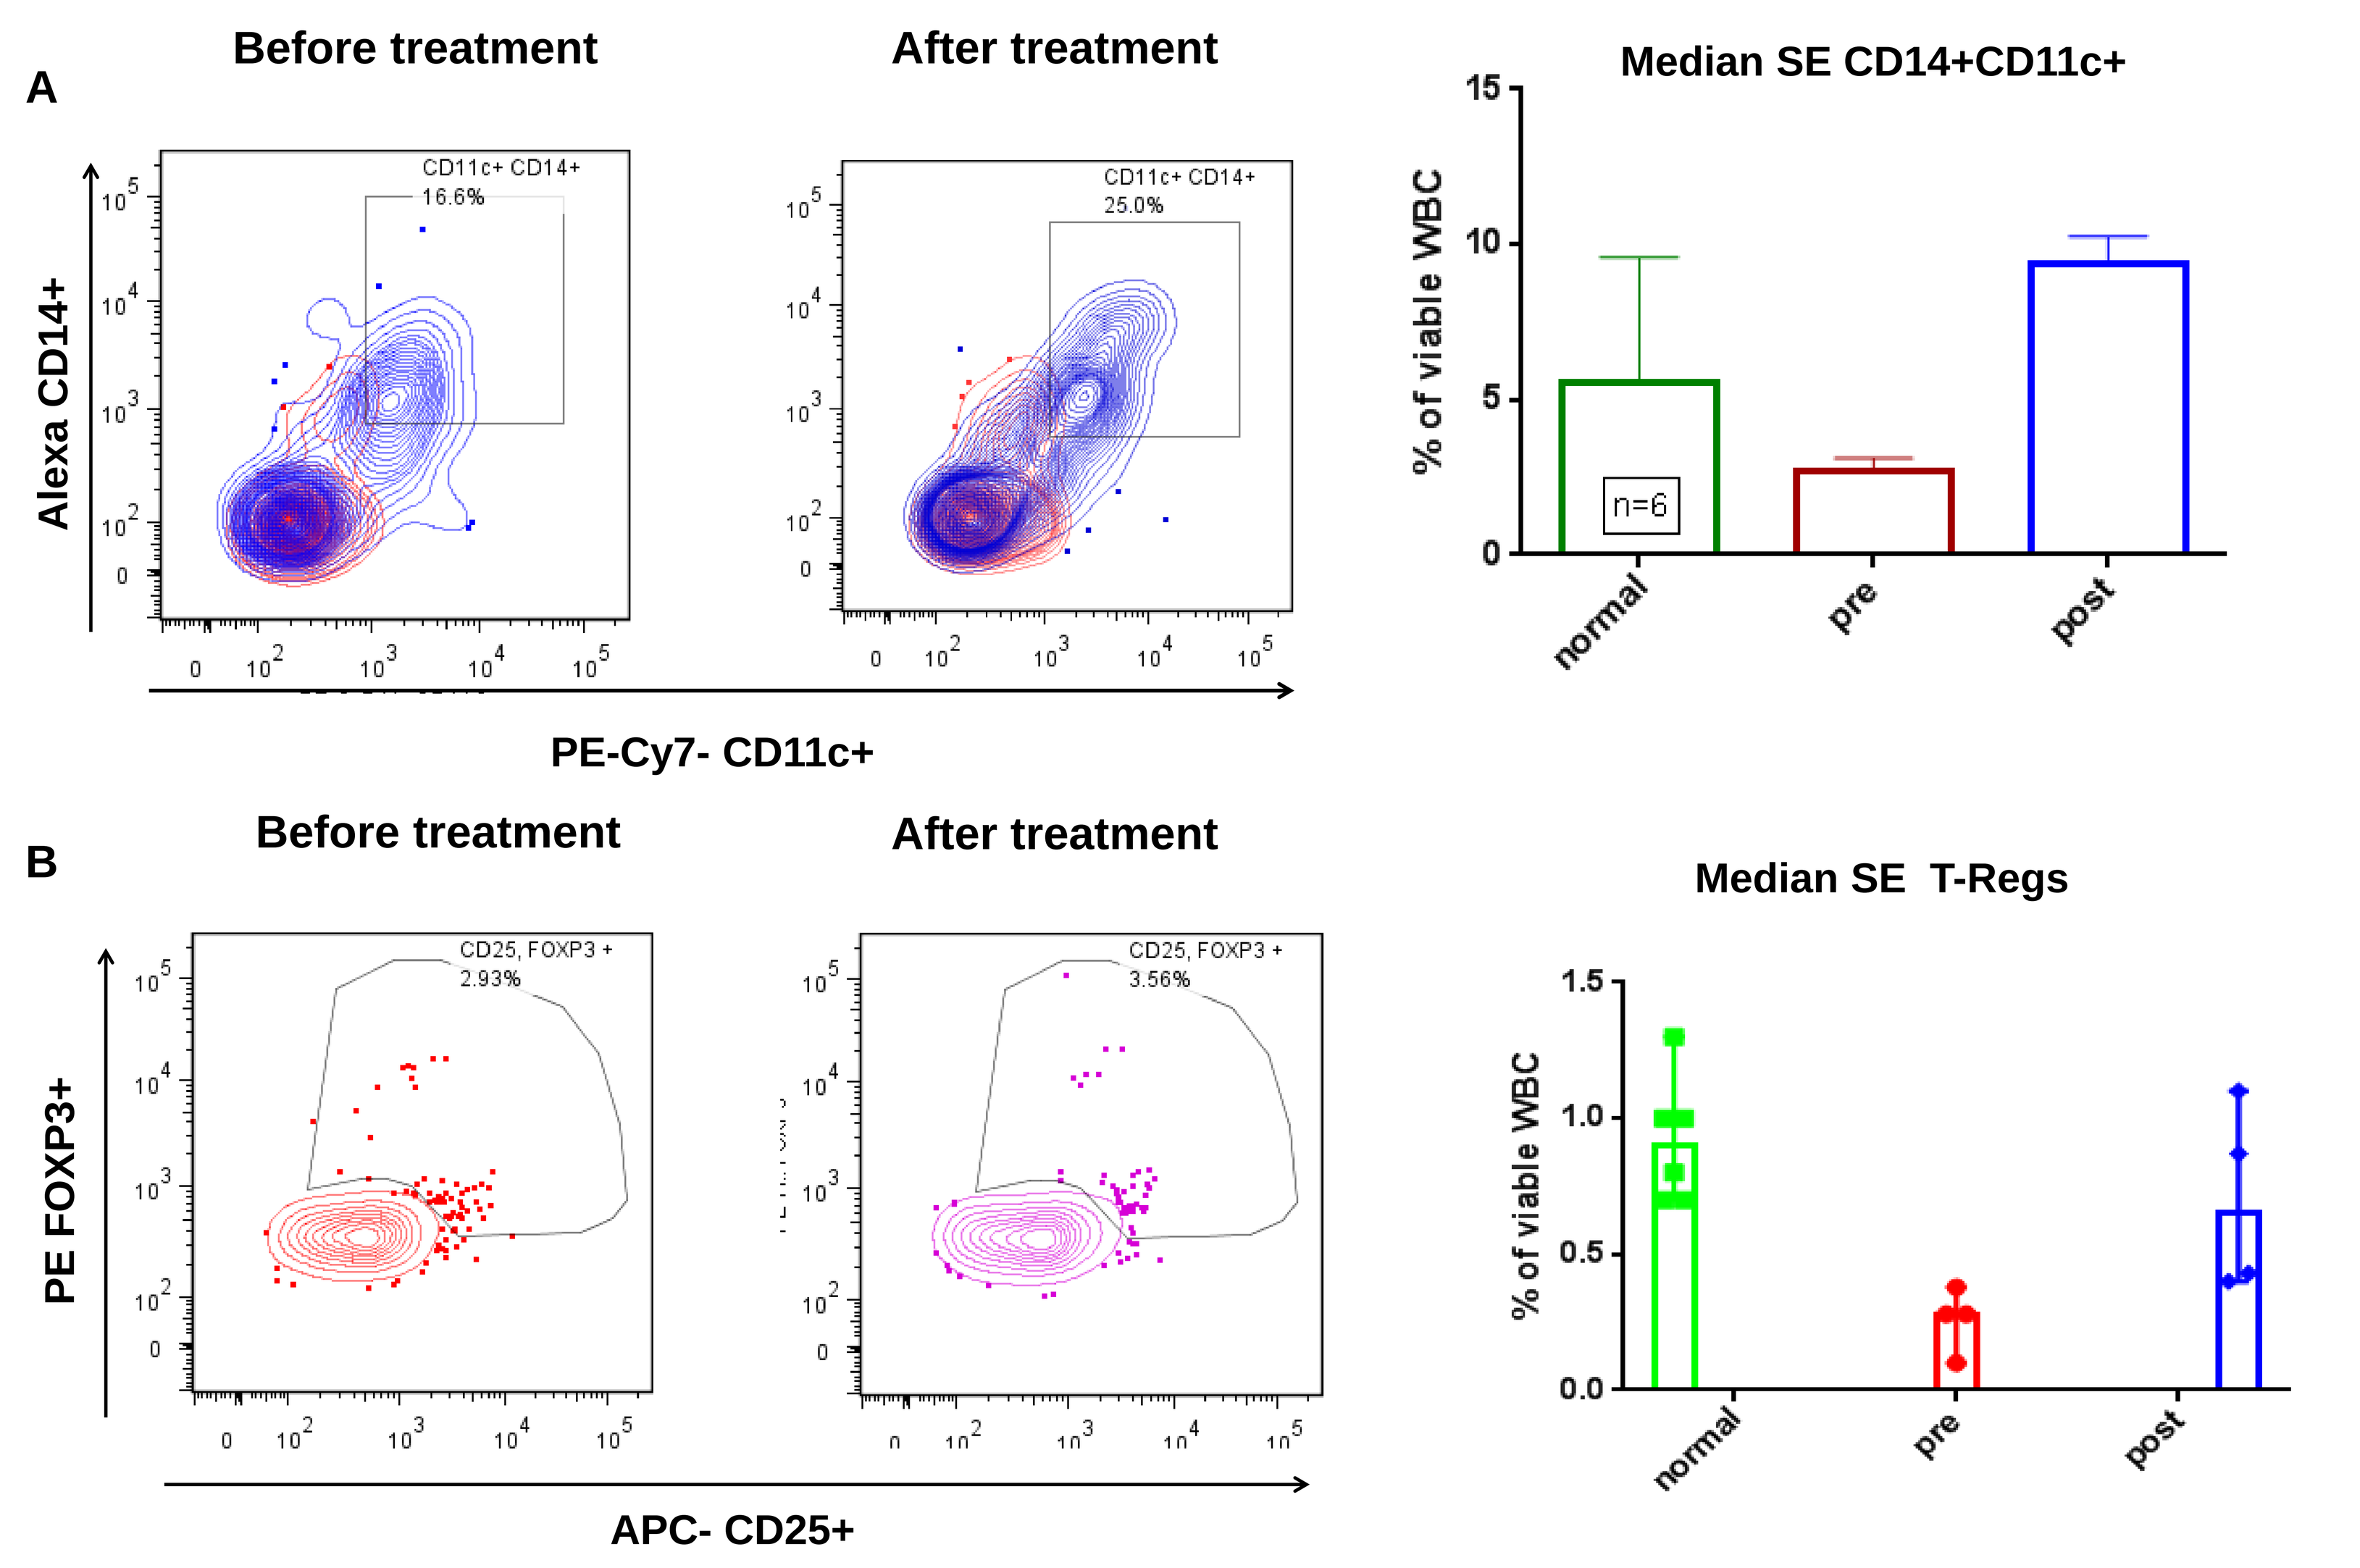

A
Before treatment
After treatment
CD11c+ CD14+
16.6%
CD11c+ CD14+
25.0%
Alexa CD14+
PE-Cy7- CD11c+
Median SE CD14+CD11c+
% of viable WBC
n=6
normal
pre
post
B
Before treatment
After treatment
CD25, FOXP3 +
2.93%
CD25, FOXP3 +
3.56%
PE FOXP3+
APC- CD25+
Median SE T-Regs
% of viable WBC
normal
pre
post

Figure 5 FACS Cellular Responses were evaluated in healthy controls and trial enrolled subjects at different times in the course of the trial. Each symbol represents an individual frequency of CD11c⁺ CD14c⁺ (dendritic cells) (**A**) and CD4⁺CD25⁺FoxP3⁺ Tregs **(B)** after AAT treatment. The bars represent the mean values (n = 7, per group).

Summary and Conclusion

- **Continuous administration of AAT as salvage therapy for steroid resistant gut GVHD is feasible without clinically toxicity.**
- **Stool showed a decrease in intestinal AAT loss with AAT treatment, as measured by AAT clearance and endoscopic evaluation suggesting healing of the bowel mucosa.**
- **AAT administration suppressed serum levels of pro-inflammatory cytokines, and interfered with GVHD manifestation.**
- **In summary, Preliminary results are encouraging, and further exploration of AAT therapy in extended phase II and randomized trials as therapy of steroid refractory acute GVHD or as first line therapy are warranted.**

This Clinical Trial was funded in part by: Baxter, Westlake Village, CA, USA, Omni Bio, Boulder, CO, USA and Kamada Ltd., Israel,